Closing the HRR Testing Gap in Advanced Prostate Cancer

David M. Waterhouse, MD, MPH, FASCO[1]; Molly Mendenhall, MBOE, BSN, LSSBB, RN[1]; Dan Davies[1]; David R. Drosick, MD[1]; Patrick Ward, MD, PhD[1]; Matthew H. Bertke, MD[1]; Mitchell Singstock, MD[2]; Suzzette Arnal, PhD[3]

[1] OHC (Oncology Hematology Care)/The US Oncology Network, Cincinnati, OH · [2] Department of Internal Medicine, University of Utah Health, Salt Lake City, UT · [3] The US Oncology Network, The Woodlands, TX

Abstract 584 · Poster Session A

2026 ASCO Quality Care Symposium

BACKGROUND

- NCCN guidelines support germline and somatic HRR testing across high-risk (HR), very high-risk (VHR), regional, and metastatic prostate cancer.[1–3] Germline testing is **"recommended"** across all cohorts; somatic testing is **"considered"** in HR/VHR disease and **"recommended"** in M1a–c disease.
- Results inform PARP-inhibitor eligibility, prognosis, treatment sequencing, and cascade testing.[1,2]
- Real-world adoption lags: only about one-third of men with metastatic disease are tested.[4,5]

METHODS

Using DMAIC and Plan-Do-Study-Act methodologies, we leveraged quality infrastructure validated in metastatic NSCLC biomarker testing and breast genetics referral.[6,7]

- **Baseline (2023–2024):** Retrospective chart review established rates of NCCN-concordant staging, germline/somatic HRR testing, and genetics referral.
- **Intervention (2025):** Following baseline assessment, we implemented a standardized Prostate Cancer Consult template and Order Set in McKesson G2 to streamline AJCC TNM staging and NCCN-concordant orders for HRR testing, imaging, laboratory studies, genetics referral, and follow-up.
- **Provider education:** At intervention launch, providers completed workflow-focused videos and 1:1 training with attestation, followed by targeted re-education.
- **Post-intervention monitoring:** Weekly automated EHR extraction and dashboards tracked template/order-set use, staging, genetics referral, and HRR testing, with real-time audit-feedback and re-education to address gaps.

RESULTS

Table 1. Eligible patients and testing by year

Year	Charts reviewed	Germline eligible	Germline tested	Somatic eligible*	Somatic tested
2023	259	194	54 (28%)	91	24 (26%)
2024	255	195	82 (42%)	86	37 (43%)
2025	177	170	107 (63%)	62	50 (81%)

**Germline testing eligibility included HRR, VHR, and M1a–c disease. Somatic testing is reported for the NCCN "recommended" M1a–c cohort; including HR/VHR ("consider") reduced the overall somatic testing rate to 57% as seen in Table 2.*

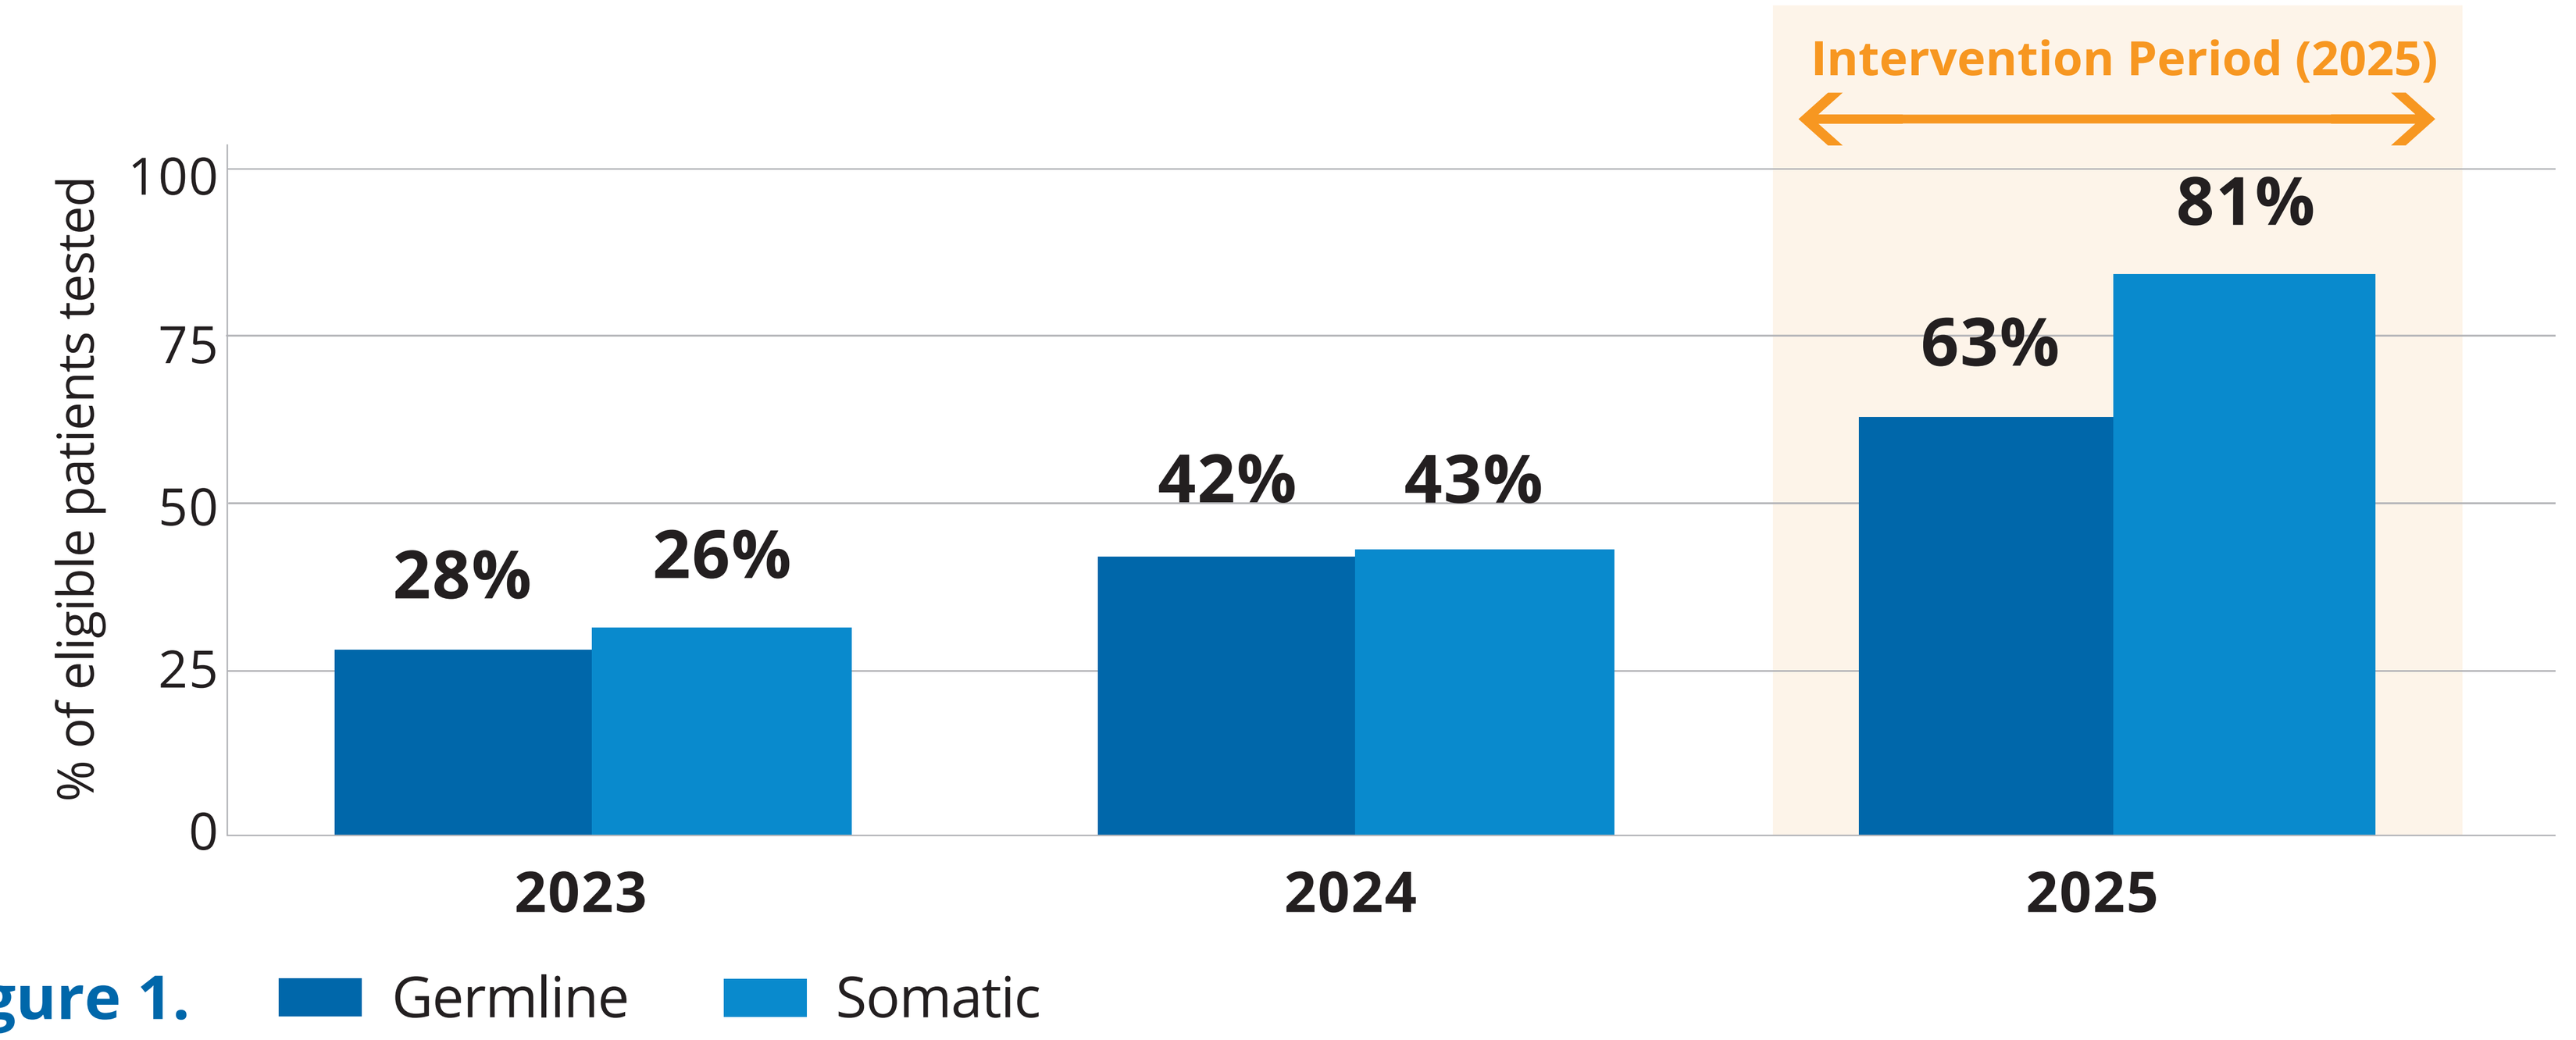
Intervention Period (2025)
% of eligible patients tested
100
75
50
25
0
28%
26%
42%
43%
63%
81%
2023
2024
2025
Germline
Somatic

Figure 1.

Audit-Feedback Closed the Gap. Can It Stay Closed?

Germline 28% → 63% Somatic 26% → 81%

The intervention required manual review and repeated outreach, limiting scalability and sustainability.

Figure 2. 2025 testing by NCCN risk cohort

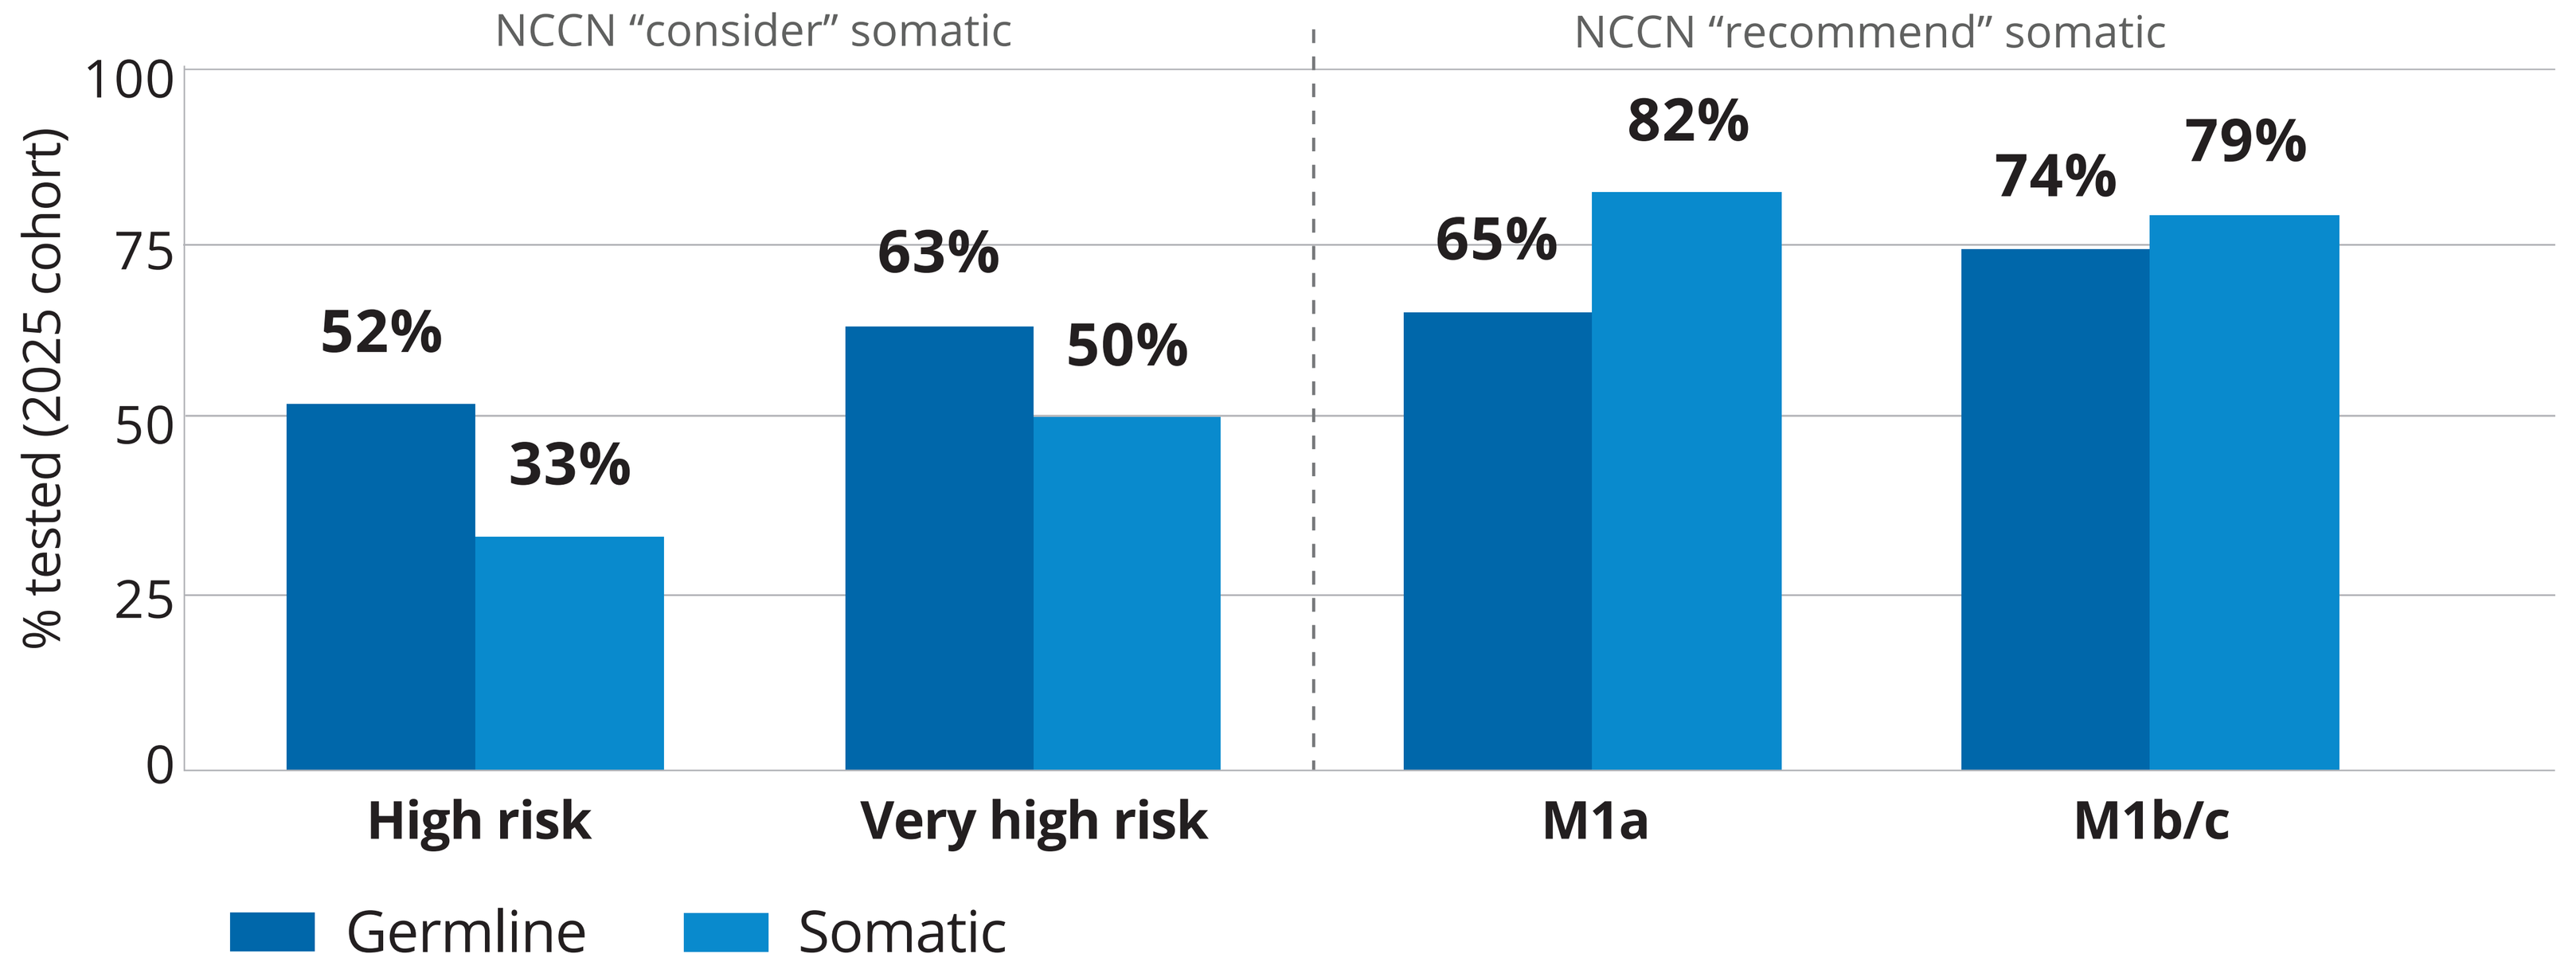
NCCN "consider" somatic
NCCN "recommend" somatic
% tested (2025 cohort)
100
75
50
25
0
52%
33%
63%
50%
65%
82%
74%
79%
High risk
Very high risk
M1a
M1b/c
Germline
Somatic

All 170 patients in the 2025 cohort. Germline testing is "recommended" across all four cohorts; somatic testing is "considered" in HR/VHR and "recommended" in M1a-c disease.

References: 1. Pritchard CC, et al. N Engl J Med. 2016;375:443-53. 2. Mateo J, et al. N Engl J Med. 2015;373:1697-708. 3. NCCN Prostate Cancer Guidelines v1.2023–v1.2025. 4. Barata PC, et al. JAMA Oncol. 2024;10:975-7. 5. Chehade CH, et al. JAMA Netw Open. 2024;7:e2431596. 6. Waterhouse D, et al. JCO Oncol Pract. 2023. 7. Waterhouse D, et al. JCO Oncol Pract. 2023. 8. Singstock M, et al. J Clin Oncol. 2026;44(16_suppl):5022

Research funding: AstraZeneca; Pfizer. **Contact: David.Waterhouse1@usoncology.com**

Figure 3. Documented reasons for non testing, 2025

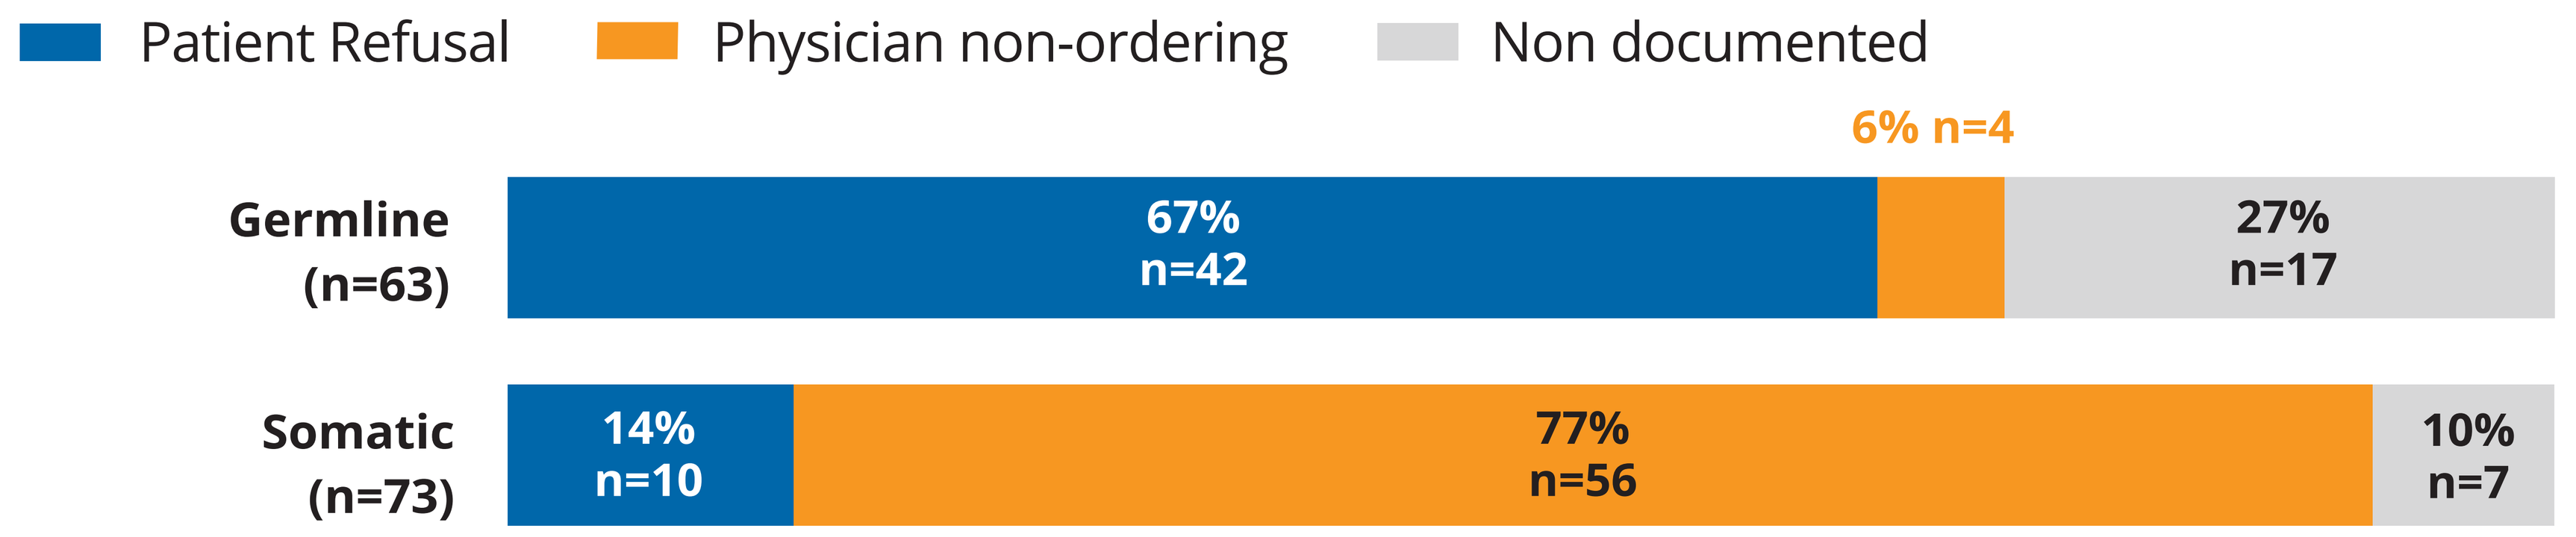
Patient Refusal
Physician non-ordering
Non documented
6% n=4
Germline (n=63)
67% n=42
27% n=17
Somatic (n=73)
14% n=10
77% n=56
10% n=7

Exploratory, not pre-specified. Germline gaps in radiation oncology–only encounters reflected lower referral and ordering intensity rather than excess patient refusal.

Table 2. 2025 testing by specialty exposure

Specialty exposure	Germline tested*	Somatic tested*
Medical + radiation oncology	16/21 (76%)	15/21 (71%)
Medical oncology only	33/52 (64%)	40/52 (77%)
Radiation oncology only	58/97 (60%)	42/97 (43%)
Overall	**107/170 (63%)**	**97/170 (57%)**

** Germline testing eligibility included HRR, VHR, and M1a-c disease. Somatic testing is reported only for the NCCN "recommended" M1a-c cohort.*

RESULTS SUMMARY

- Testing improved 2023 → 2025: germline 28% → 42% → 63%; somatic 26% → 43% → 81% (both $p < 0.001$).
- Following intervention, 2025 germline testing remained suboptimal despite NCCN recommendation: 52% HR, 63% VHR, 65% M1a, 74% M1b/c.
- 2025 Somatic testing mirrored NCCN wording — lower where "considered" (HR 33%, VHR 50%), higher where "recommended" (M1a 82%, M1b/c 79%).
- Germline gaps reflected patient refusal (42/63, 67%); somatic gaps, physician non-ordering (56/73, 77%).

CONCLUSIONS

- A template, order set, and audit-feedback substantially improved germline and somatic HRR testing, but gaps persisted — particularly in HR and VHR disease.
- Somatic testing was lower among patients seen only in radiation oncology.
- Reliance on manual abstraction and repeated outreach limits sustainability and scalability.
- **Building on our prior work demonstrating AI accuracy in prostate cancer staging, risk stratification, and NCCN-concordant testing assignment (ASCO 2026 Abstract 5022, #535732),[7] these findings support future agentic AI–enabled case identification and provider prompting within a human-in-the-loop workflow.**